Comparison of Inter-Occasion and Inter-Individual Variability in Chemotherapy - Induced Myelosuppression

UPPSALA UNIVERSITET

Emma K. Hansson(1), Johan Wallin(1), Marie Sandström(1,2), Mats O. Karlsson(1), Lena E. Friberg(1)

[1]Department of Pharmaceutical Biosciences, Division of Pharmacokinetics and Drug Therapy, Uppsala University, Uppsala Sweden,
[2]AstraZeneca R&D, Södertälje, Sweden

Background and Objectives

A semi-physiological model of chemotherapy-induced myelosuppression (Fig 1.) has previously been developed and applied to several different anticancer drugs. Consistency in system-related parameter estimates and inter-individual variability (IIV) have been reported across drug [1]. A requirement for the model to be a useful tool for individual dose adjustments based on neutrophil counts [2, 3] is relatively low variability between treatment courses (IOV) in relation to IIV.

The aim of this study was to evaluate and compare magnitudes of IOV and IIV in myelosuppression model parameters across six different data sets.

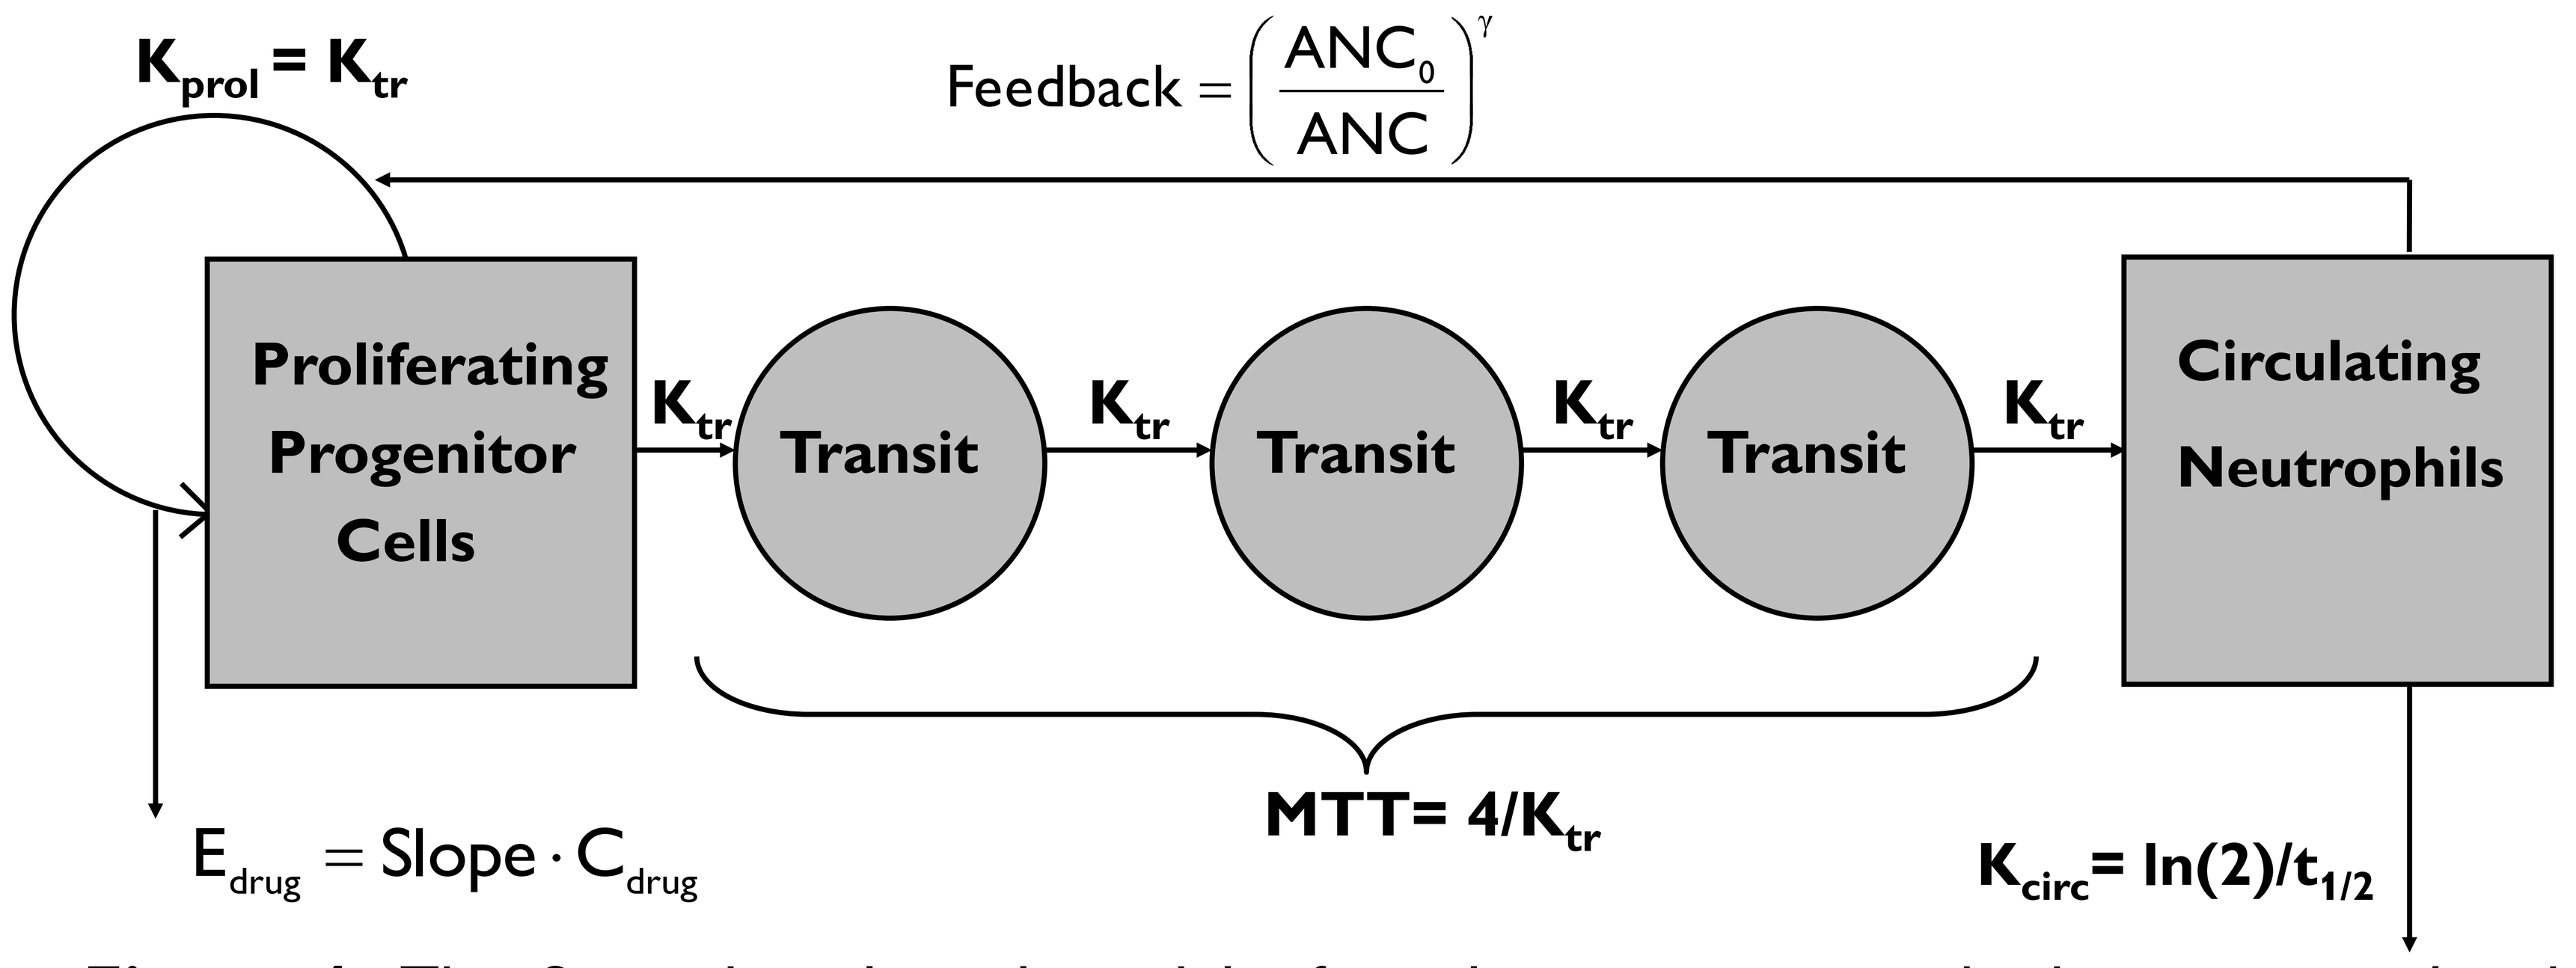
$K_{prol} = K_{tr}$
$\text{Feedback} = \left(\frac{ANC_0}{ANC}\right)^{\gamma}$
Proliferating Progenitor Cells
K_{tr}
Transit
K_{tr}
Transit
K_{tr}
Transit
K_{tr}
Circulating Neutrophils
$E_{drug} = \text{Slope} \cdot C_{drug}$
MTT= 4/K_{tr}
K_{circ}= ln(2)/$t_{1/2}$

Figure 1. The Semi-physiological model of myelosuppression with the system related parameters baseline neutrophil count (ANC_0), mean transit time (MTT), feedback factor γ and the drug effect parameter Slope estimated. $t_{1/2}$= 7 hours

Methods

Neutrophil counts from several treatment courses were available following therapy with docetaxel, paclitaxel, epirubicin-docetaxel, 5-fluorouracil-epirubicin-cyclophoshamide, topotecan and etoposide. One occasion was defined as one treatment course.

The PK of the drugs were described using individual PK parameters from previously determined PK models [4, 5, 6, 7, 8] or population PK parameters [9]. IOV in PK was not available.

The semi-physiological myelosuppression model [1] was fitted to the neutrophil observations using the FOCE method in NONMEM VI. The subroutine PRIOR was used to estimate separate Slope parameters for the co-administered drugs. The data were Box-Cox transformed with a factor 0.2 and the half-life of neutrophils was fixed to 7 hours.

IOV in baseline neutrophil count (ANC_0), mean transit time (MTT) and Slope were evaluated for statistical significance ($P < 0.001$).

Results

Table 1. Typical population parameter estimates and residual error (relative SE %)

Data Set	ANC_0 (x 10^9/L)	MTT (hours)	$Slope_1$ (L/mg)	$SLope_2$ (L/mg)	γ	Residual Error †
Docetaxel	4.81 (2.6)	94.0 (1.6)	21.4 (3.4)	-	0.17 (1.8)	0.53 (1.1)
Paclitaxel	5.61 (9.4)	154 (4.4)	81.5 (8.1)	-	0.27 (5.9)	0.43 (2.6)
Epirubicin-docetaxel	3.49 (11)	117 (3.1)	32.1 (29)*	21.8 (32)**	0.21 (5.1)	0.50 (3.4)
5-Fu-epirubicin-cyclophosphamide	4.56 (5.3)	184 (3.2)	32.2 (47)*	26.6 (22)***	0.24 (2.4)	0.54 (1.9)
Topotecan	7.11 (8.8)	159 (5.7)	0.0715 (23)	-	0.27 (7.9)	0.45 (1.1)
Etoposide	5.67 (9.9)	165 (6.3)	0.213 (12)	-	0.14 (3.4)	0.49 (4.3)

† On Box-Cox transformed scale

*Epirubicin **Docetaxel ***Cyclophosphamide

Table 2. Estimated IIV CV %, IOV CV % (relative SE %) and Δ reduction in residual error %

Data set	IIV ANC_0	IIV MTT	IIV Slope	IOV ANC_0	IOV MTT	IOV Slope	Δ Residual Error %
Docetaxel	33 (5.9)	9.0 (19)	37 (7.0)	-	16 (4.8)	19 (12)	9.1
Paclitaxel	36 (13)	17 (22)	39 (20)	-	16 (8.5)	-	23
Epirubicin-docetaxel	37 (15)	13 (21)	22 (23)	-	8.0 (20)	-	8.9
5-Fu-epirubicin-cyclophosphamide	28 (15)	16 (13)	23 (14)	-	7.5 (11)	-	7.0
Topotecan	28 (27)	17 (33)	61 (37)	15 (39)	-	29 (19)	7.4
Etoposide	47 (15)	24 (25)	32 (52)	-	13 (35)	40 (25)	23

The myelosuppression model could well characterize the neutrophil-time course and resulted in similar system-related parameter estimates as previously observed (Table 1) [1].

IOV in MTT was significant and limited for all the investigated datasets except for topotecan (Table 2). For docetaxel and etoposide IOV in Slope was also found significant. For topotecan IOV in Slope and ANC_0 was significant. The estimated overall IOV were clearly lower than IIV in all cases (Figure 2). By inclusion of IOV the residual errors decreased on average by 13%.

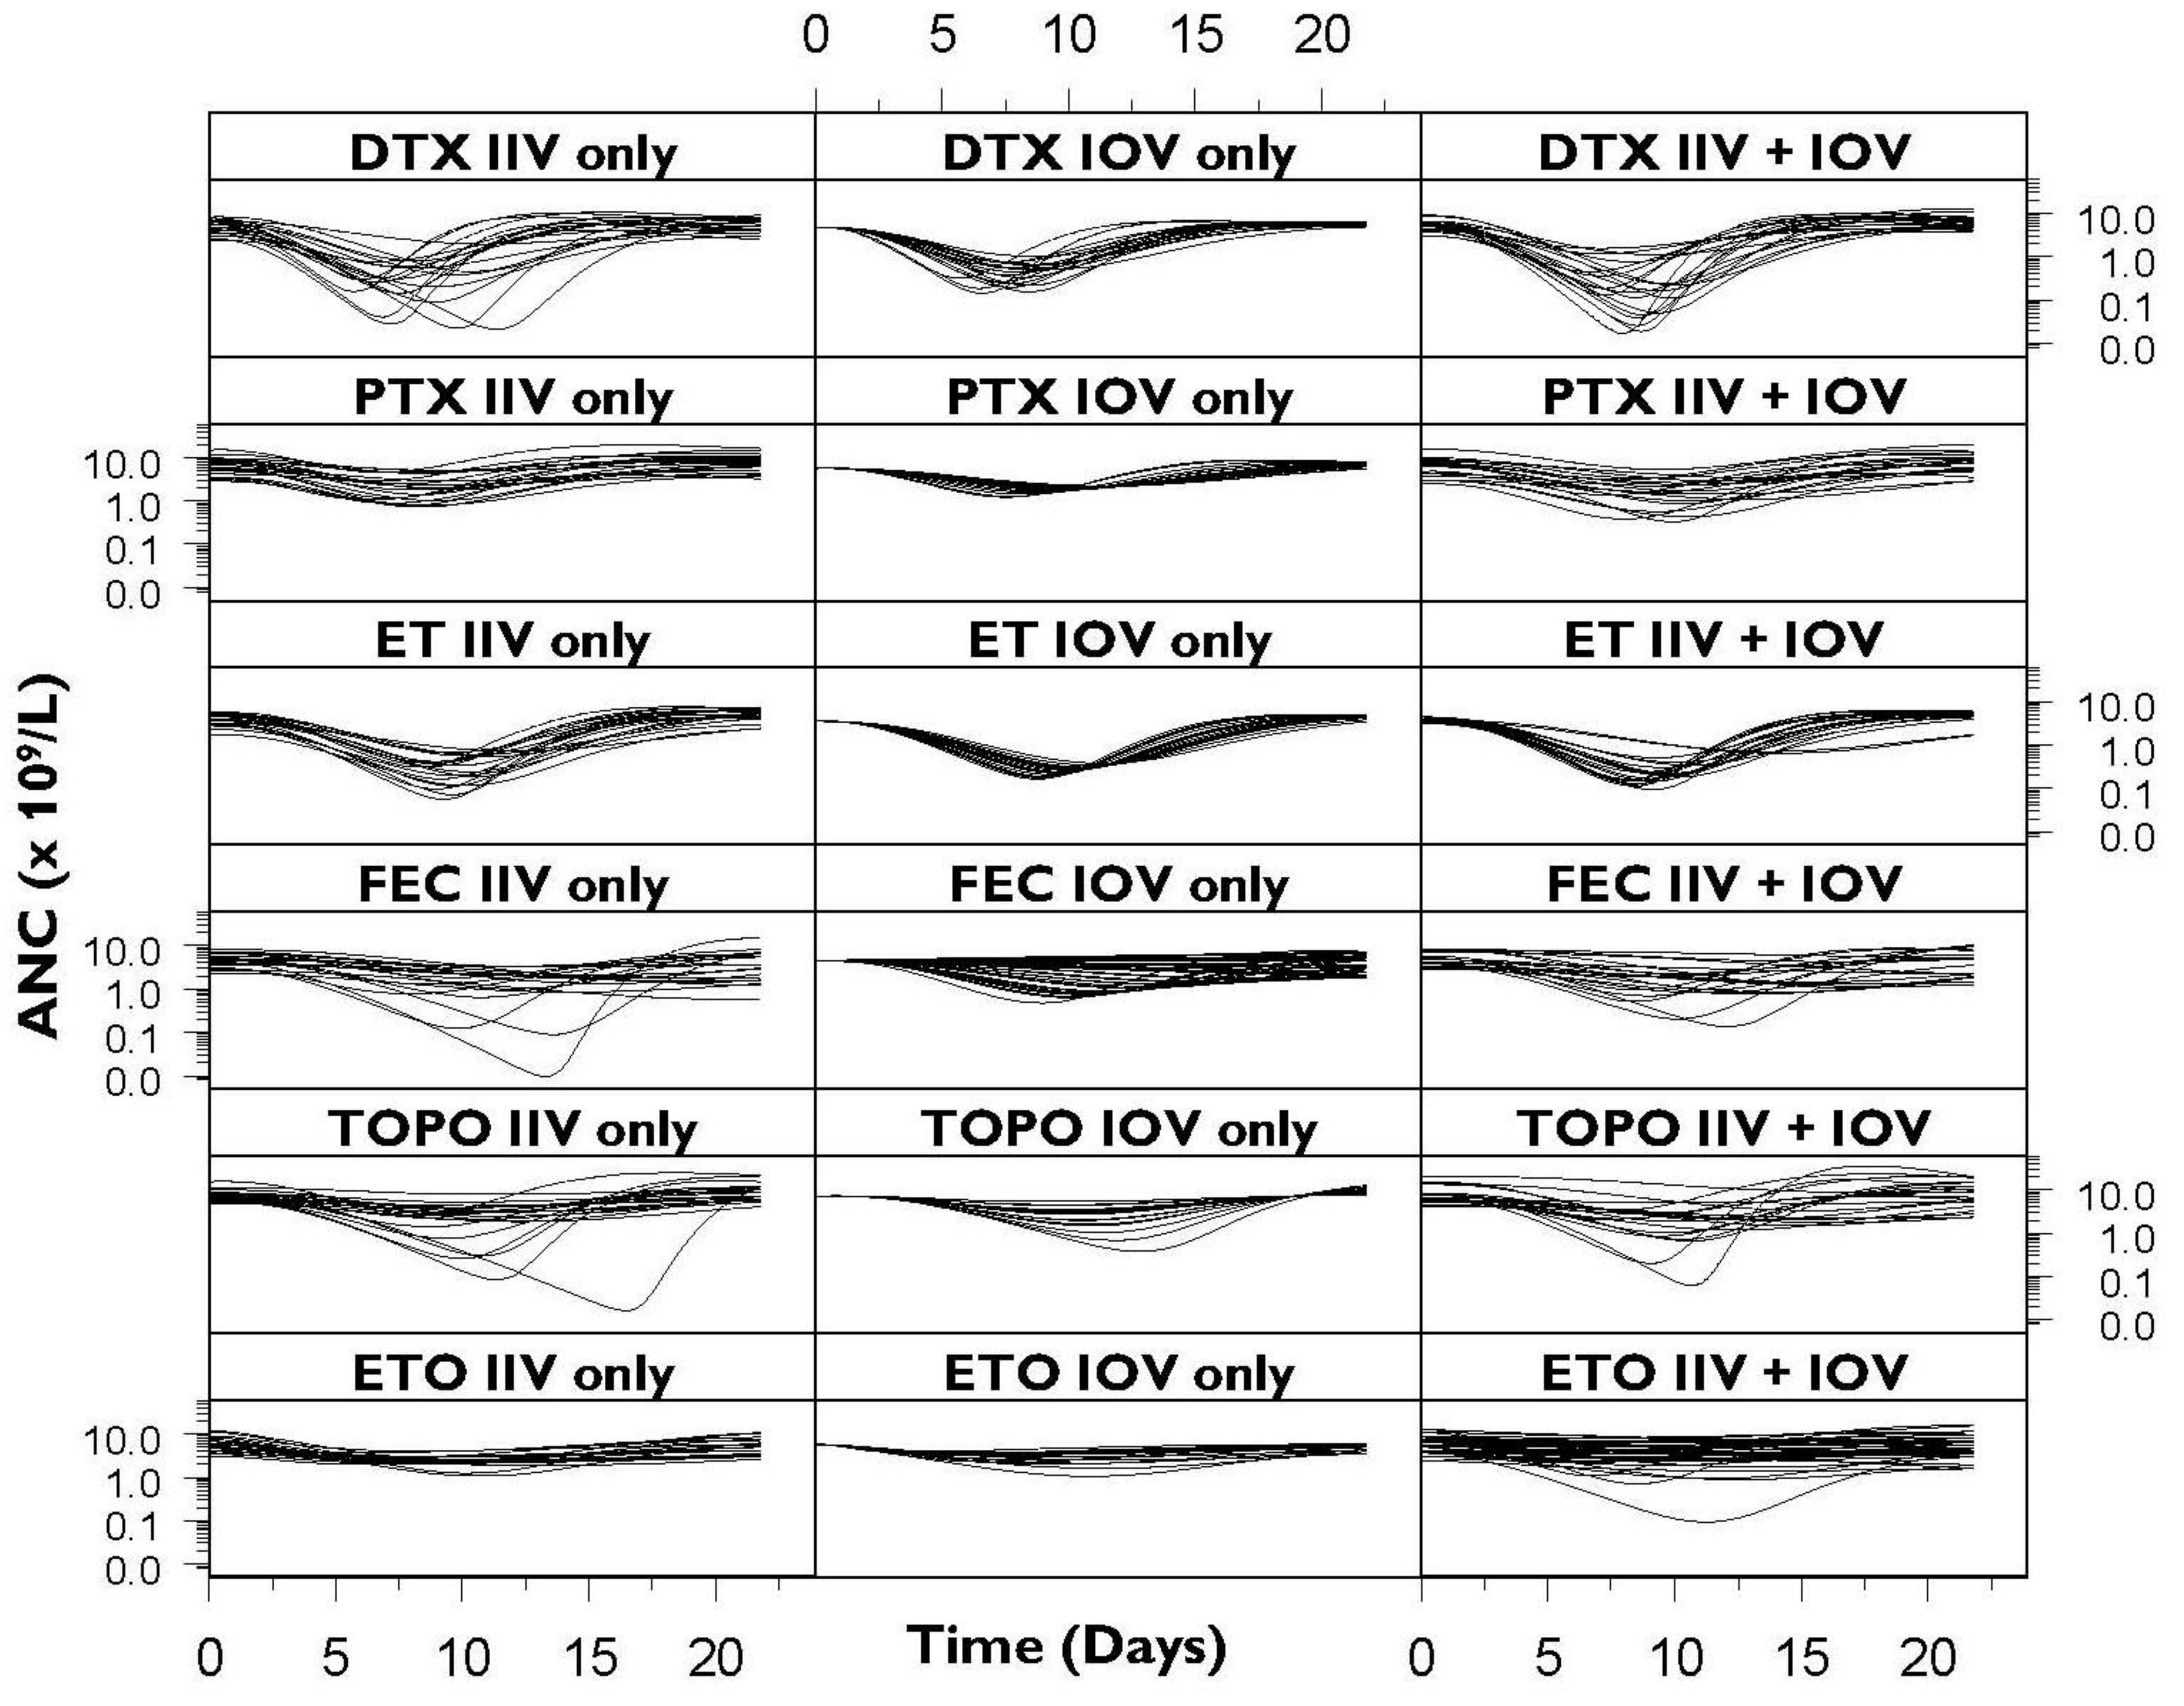
DTX IIV only
DTX IOV only
DTX IIV + IOV
PTX IIV only
PTX IOV only
PTX IIV + IOV
ET IIV only
ET IOV only
ET IIV + IOV
FEC IIV only
FEC IOV only
FEC IIV + IOV
TOPO IIV only
TOPO IOV only
TOPO IIV + IOV
ETO IIV only
ETO IOV only
ETO IIV + IOV
ANC (x 10^9/L)
Time (Days)
10.0
1.0
0.1
0.0
0 5 10 15 20

Figure 2. Twenty simulated time-courses of myelosuppression including IIV only, IOV only or both IIV and IOV for all the six investigated datasets.

Conclusion

For all six investigated datasets the overall IOV was estimated to be lower than the overall IIV. The limited IOV in relation to IIV in the myelosuppression model parameters indicate that the semi-physiological model has potential as a tool for individual dose adjustment based on neutrophil counts for which a tool is under development [2].

References

[1] Friberg LE., et al., J Clin Oncol; 20:4713-21, 2002.
[2] Wallin J., et al., PAGE 15 (2006) Abstr 966 [www.page-meeting.org/?abstract=966].
[3] Wallin J., et al., PAGE 15 (2007) Abstr 1169 [www.page-meeting.org/?abstract=1169.
[4] Henningsson A., et al., Eur J Cancer; 39:1105-1114, 2003.
[5] Sandström M., et al., J Clin Oncol; 23:413-21, 2005.
[6] Sandström M., et al., J Clin Oncol; 58:143-56, 2006.
[7] Toffoli G., et al., J Clin Pharmacol; 52:511-519, 2001.
[8] Legér F., et al., British J Cancer 90:343-347, 2004.
[9] Bruno R., et al., J Pharmacokinet Biopharm; 24:153-72, 1996.

Acknowledgment: We would like to thank F. Hoffman La-Roche ltd. for kindly providing the docetaxel data.